INNOVATION, R&D - Laboratoires Expanscience

Early Atopic Dermatitis biological analysis

Clarence de Belilovsky[1], Gaëtan Boyer[1], A. Moga[2], M. Fontanié[3], Caroline Baudouin[1], Gaëlle Bellemère[1]

1 Innovation R&D Direction, Laboratoires Expanscience, Epernon, France 2 Synelvia, Labège, France 3 Vibiosphen, Labège, France

Background

Pathophysiology of atopic dermatitis (AD) is characterized by decreased levels of Natural Moisturizing Factors (NMFs) and ceramides, with microbiota dysbiosis (colonization by staphylococci species). Little is known about microbiota, NMFs and ceramides before the onset of Atopic Dermatitis (AD) and during first AD flare in children with and without a genetic risk of AD.

Objectives

The main objective was to quantify NMFs, ceramides, Staphylococcus aureus (SA) and Staphylococcus epidermidis (SE) colonization from birth until first AD flare. The impact of daily applications of emollients has been reported.

Methods

Swabs were taken on forearms, face and AD lesions in 45 infants for staphylococci, 76 for NMF/ceramides out of 180 newborns involved in a 6-month AD prevention study. This was an open label, randomized, multicentric, controlled trial conducted in private practice (2 pediatricians and 9 dermatologists). Children were enrolled at 2-3 weeks of life and followed at D0, D30, D90, D120, D180 with daily emollient applications (STELATOPIA®) (Graph 1). Children were analyzed for SA and SE by quantitative PCR and for NMF and ceramides by LC/UV and LC/MS (Graph 2). D-squames were performed on forearms of 6 children to visualize SA and its biofilm if any using Electron Microscopy.

Graph 1: Clinical study flow chart

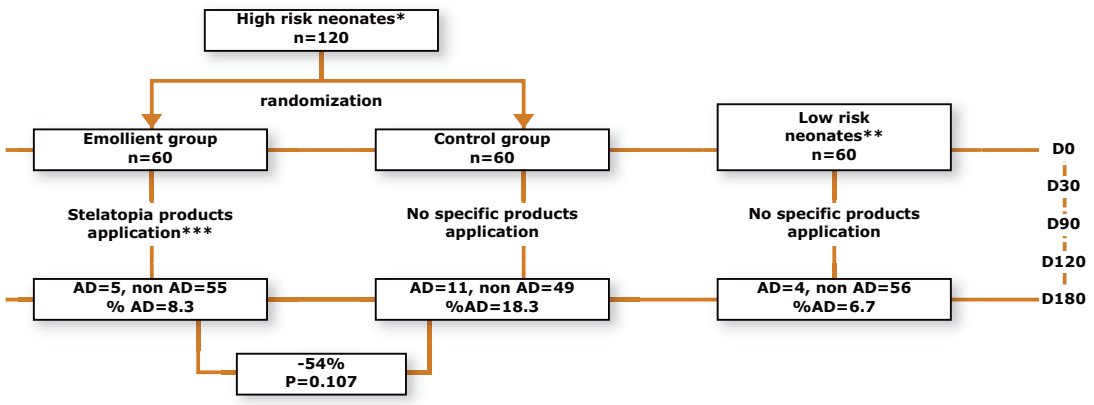
High risk neonates*
n=120
randomization
Emollient group
n=60
Control group
n=60
Low risk
neonates**
n=60
Stelatopia products
application***
No specific products
application
No specific products
application
AD=5, non AD=55
% AD=8.3
AD=11, non AD=49
%AD=18.3
AD=4, non AD=56
%AD=6.7
-54%
P=0.107
D0
D30
D90
D120
D180

*High risk neonates: 2 parental history of atopy
**Low risk neonates: non parental history of atopy
***Stelatopia Emollient balm, twice a day + Stelatopia Cleansing cream, twice a week + Stelatopia Bath Oil, twice a week

Graph 1: Clinical study flow chart

	TOTAL	High risk				Low risk
	45	Emollient		Control		
		AD (5)	No AD (10)	AD (10)	No AD (10)	No AD (10)
S aureus						
Forearm	6/45 (13.3%)	1	1	1	1	2
Face	15/41 (36.6%)	1	3	2	2	7
AD lesion	2/11 (18.1%)	0/4	2/7			
S epidermidis						
Forearm	42/45 (93.3%)	4	10	10	10	8
Face	35/41 (85.4%)	3	10	5	10	7
AD lesion	9/11 (81.8%)	3/4	6/7			

Results

This study is a AD prevention study using emollients from birth demonstrated clinical efficacy, with a 54% decrease of AD occurrence at 6 months of life (D180).
Pooled results from D0 (2-3 weeks of life) to D180 showed that infants were poorly colonized with SA: only 6 children among 45 on forearms and 15 on the face. In contrast, infants were largely colonized with SE: 42 infants on the forearm and 35 on the face. Only 2 AD lesions out of 11 were found positive for SA and 9 for SE. Levels of SA and SE colonization were not modified by emollients. Only one infant presented SA biofilm at D0 (Photos 1). SA biofilm tended to disappear in the course. As this child was in the emollient group we could follow the diminution and disappearance both of SA and biofilm with time (Photos 2 and 3).

Photos 1 (X5000): Staphylococcus aureus biofilm at D0

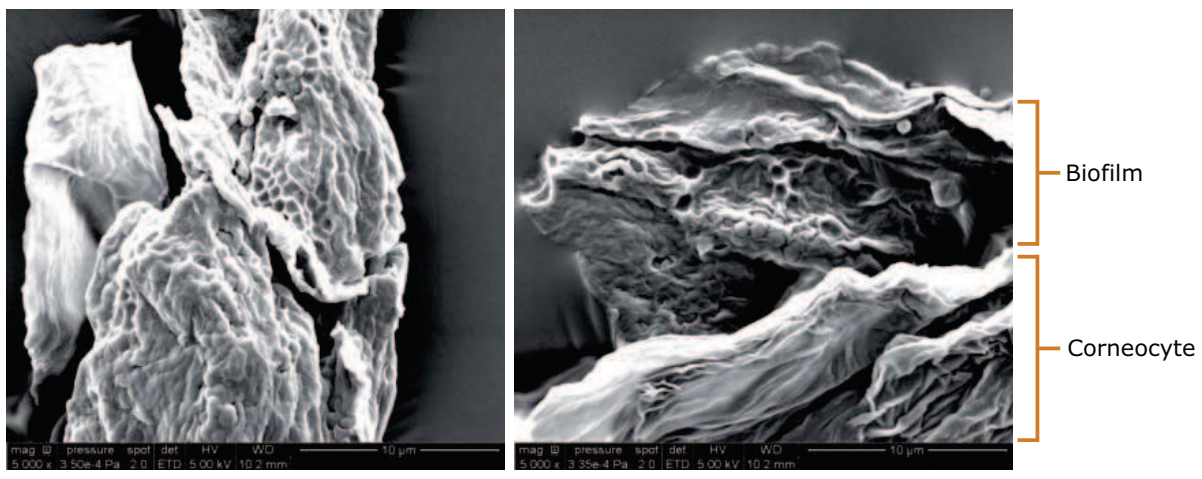
Biofilm
Corneocyte

Photos 2 (X3000) and 3 (X 1000): Staphylococcus aureus at D30 and D90

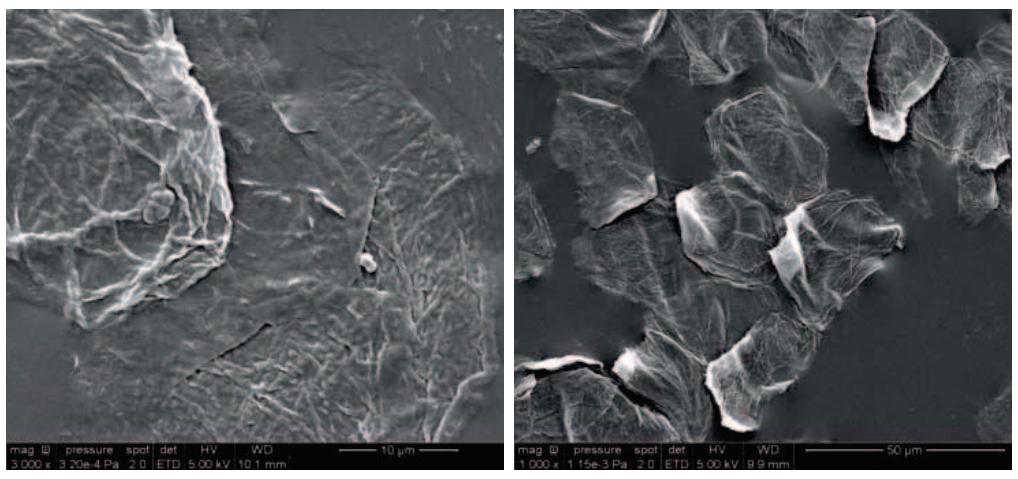

At D0 (2-3 weeks of life) a global depletion of NMFs has been observed on forearms in infants at high risk of developing AD as compared to infants at low risk (Graph 3 a)). In contrast no statistical difference has been observed in NMFs levels at D0 between infants with high risk that developed AD and those that did not develop AD (Graph 3 b)).

The depletion of NMFs in high risk infants has been confirmed at all time points up to D180 (Graph 4). Interestingly, daily application of emollients restored NMFs levels in these subjects at D180 (Graph 4).

Globally, it has been noticed that the levels of NMFs were lower in the first lesions of AD, even in those infants for which emollient products were applied daily (Graph 4). The levels of ceramides were not different between children at high risk and at low risk of AD nor between AD and non AD infants at D0 (Graph 5).

Similarly, at each visit, no significant difference has been observed between infants at high risk of developing AD and infants with no identified risk (Graph 6).

Application of Stelatopia emollient products globally increased the levels of ceramides in high risk or in low risk group, with a statistical difference at D180 (Graph 6). Globally, it has been noticed that the levels of Ceramides were lower in the first lesions of AD, even in those infants for which emollient products were applied daily (Graph 6).

Graph 3: Quantification of NMFs at D0.
Comparison between a) high risk and low risk subjects and
b) subjects from high risk groups developing future and those without future AD
**p<0,1*

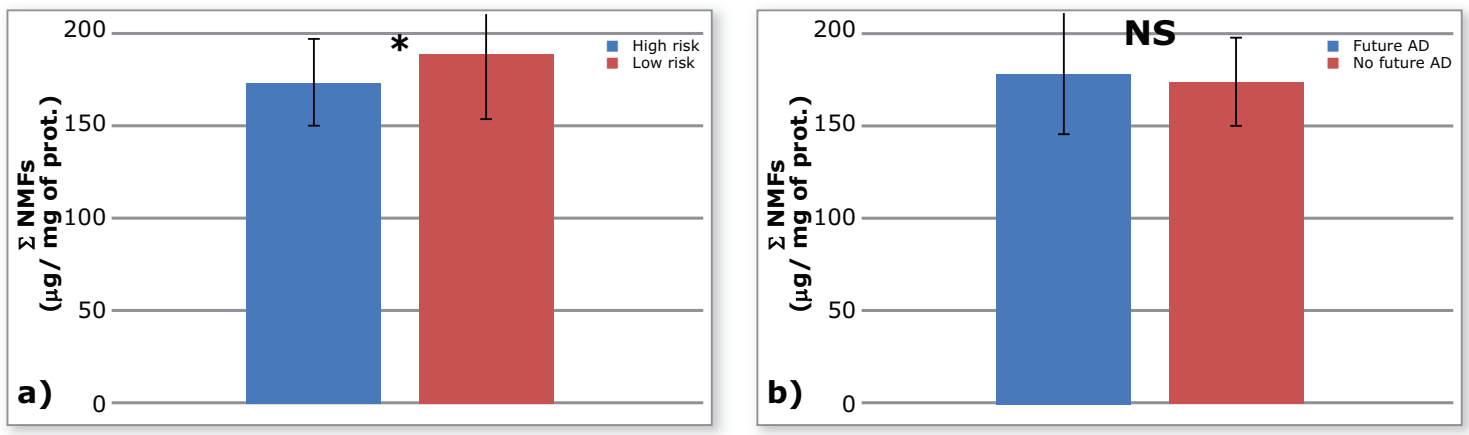
200
150
100
50
0
Σ NMFs
(µg/ mg of prot.)
*
High risk
Low risk
a)
NS
Future AD
No future AD
b)

Graph 4: Quantification of NMFs on forearm for each subject group at each time point (solid line, mean group values) and in first AD lesions (cross, individual datas)
**p<0,1*

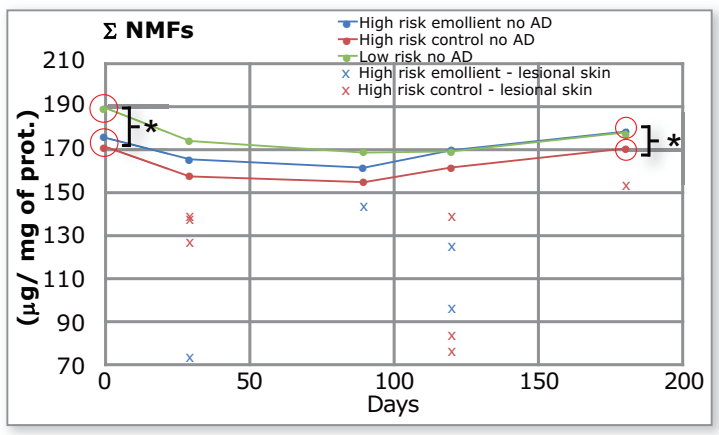
Σ NMFs
High risk emollient no AD
High risk control no AD
Low risk no AD
High risk emollient - lesional skin
High risk control - lesional skin
210
190
170
150
130
110
90
70
(µg/ mg of prot.)
0
50
100
150
200
Days
*
*

Graph 5: Quantification of Ceramides at D0.
Comparison between a) high risk and low risk subjects and
b) subjects from high risk groups developing future and those without future AD

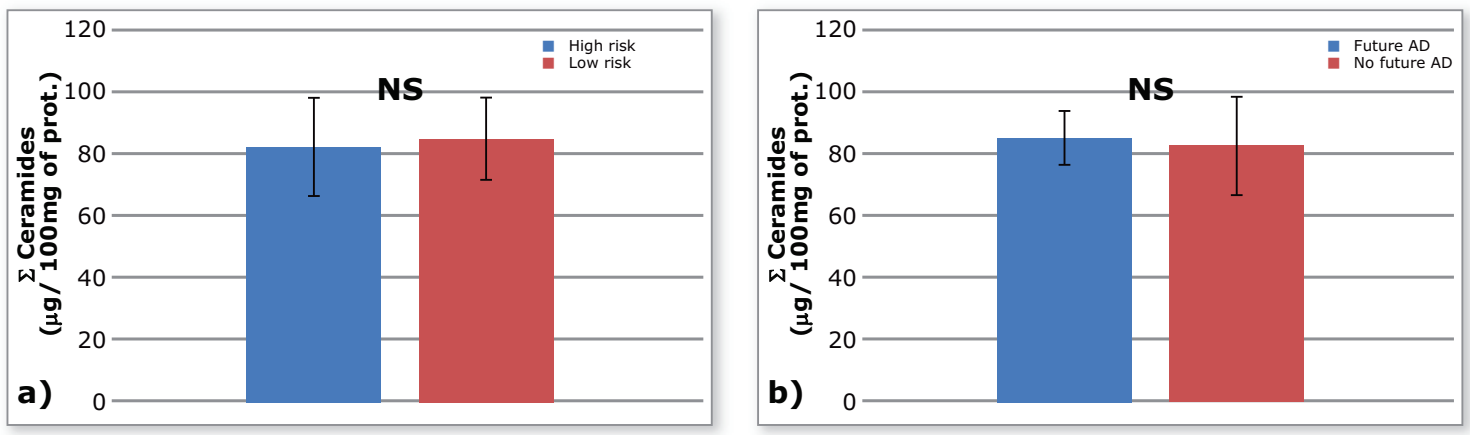
120
100
80
60
40
20
0
Σ Ceramides
(µg/ 100mg of prot.)
High risk
Low risk
NS
a)
Future AD
No future AD
NS
b)

Graph 6: Quantification of Ceramides on forearm for each subject group at each time point (solid line, mean group values) and in first AD lesions (cross, individual datas)
***p<0.05*

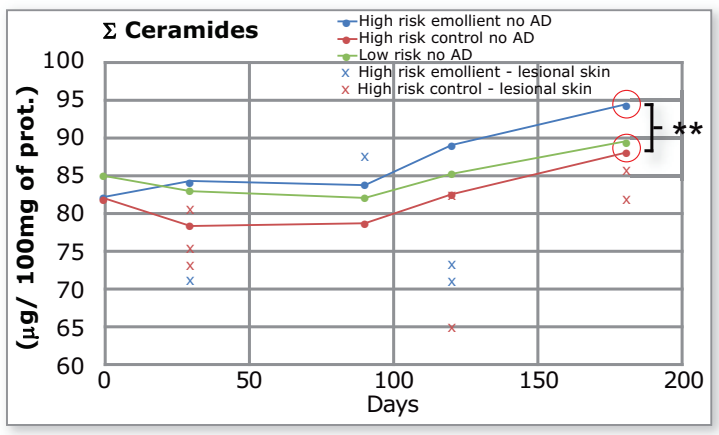
Σ Ceramides
High risk emollient no AD
High risk control no AD
Low risk no AD
High risk emollient - lesional skin
High risk control - lesional skin
100
95
90
85
80
75
70
65
60
(µg/ 100mg of prot.)
0
50
100
150
200
Days
**

Discussion

AD prevention is becoming a major challenge. Simpson (2014) and Horimukai (2014) opened the way to future large prevention clinical trials. Our prevention study in neonates with a high genetic risk of developing AD (2 or more parents or siblings with atopy) demonstrated that the application of an emollient specifically designed to prevent AD and used routinely with cleansing gel and bath oil for 6 months, lead to a decrease of 54% of the relative risk to develop AD in neonates.
To better improve prevention program and skin cares for early AD in infants, it is crucial to consider the impact of genetic background in AD, to describe clinical and biological predictive signs of AD and to understand the physiopathology of the lesions during the first flares. Increased TEWL was already identified as a risk factor in AD: increased TEWL was measured on forearms at 2 days of age in newborns that developed AD at 1 year of age (Kelleher, 2015) and for food allergy at 2 years of age (Kelleher, 2016). Increased TEWL was also measured on the front during the first week of life in infants that developed AD at 32 weeks of age. Genetic background is the most prominent risk factor identified so far and family history of AD is today the main criterion used to enroll infants in prevention program.
Our results demonstrated that the skin was poorly colonized with SA prior to AD and during the first AD flare. These data are different from the data obtained in adult AD patients showing low microbial diversity and dominance of SA. Nevertheless our results are in good agreement with the recent publication from Kennedy et al (2017) showing that patients with infantile AD did not present noticeably dysbiotic microbial communities before the onset of AD or during the first flare, and that these patients were not colonized by SA.
It should be noted that commensal staphylococci were significantly less abundant in infants affected at month 12, suggesting that this genus might prevent later development of AD. Interestingly, emollients used in the present study could maintain the high level of SE suggesting that this product is of clinical relevance in AD prevention program.
In child and adult AD, biofilm of SA is often observed (Masako et al, 2005; Gupta et al, 2016) and is supposed to play a role in chronic inflammation and pruritus (Allen et al, 2014). In our study visualization of skin surface in D-Squame samples by electron microscopy has revealed that only one neonate presented visible biofilm on superficial skin. Presence and involvement of SA biofilm in AD physiopathology in infants warrants larger studies and further investigation.
Natural moisturizing factors are degradation products of filaggrin (FLG) and low levels of NMFs have been described in AD patients carrying FLG mutations (mostly moderate and severe AD). A depletion of NMFs is correlated with dry skin and is observed in all types of AD, but it appeared to be higher in case of FLG mutation (Mlitz et al, 2012). The present study demonstrated that NMFs levels are significantly lower in newborns with family history of AD (high risk babies) but no difference has been observed in newborns that will develop AD suggesting that if atopic background is a risk factor for NMF depletion after birth no direct correlation could be made with early AD. Specific emollient could restore NMFs levels and we may speculate that this is one of their mode of action for AD prevention.
Previous study has shown that ceramide levels are decreased during AD in adults. We did not measure any modification in the levels of superficial ceramides before the onset of AD or during first AD flare in infants. Similarly, no difference in ceramide levels has been measure on superficial skin between infants at high risk and those at low risk. The involvement of ceramides in the onset and the physiopathology of infantile AD in newborns requires further investigations.

Conclusion

This clinical study confirmed that application of AD skin care from birth on neonates with genetic risk of AD is able to decrease the occurrence of AD at 6 months of age by 54%. It provided new insights on skin microbial colonization, and on NMFs and ceramide involvement in infantile AD. Specific AD emollients improved NMFs and ceramides contents and did not alter skin microbiota.

mustela®
A brand from Laboratoires Expanscience

EXPANSCIENCE®
LABORATOIRES

"Expanscience member of the UEBT for promoting biodiversity and complying with fair trade regulations"